Modification of cortical oscillatory activity following botulinum toxin injection in post-stroke patients

Alexandre Chalard[1,2], Joseph Tisseyre[1], Philippe Marque[1,3], David Amarantini[1], David Gasq[1,4]

[1]Toulouse NeuroImaging Center, Toulouse, France; [2]Ipsen Innovation, Les Ulis, France; [3]Department of Neurological Rehabilitation, University Hospital of Toulouse, Hôpital de Rangueil, Toulouse, France; [4]Department of Functional Physiological Explorations, University Hospital of Toulouse, Hôpital de Rangueil, Toulouse, France

BACKGROUND

- Intramuscular botulinum toxin A injection is first-line treatment for muscle overactivity [1].
- Botulinum toxin A can modulate sensorimotor brain activity [2].
- Following botulinum toxin A injection, sensorimotor brain activity tends to decrease by a modulation of the afferent input [2].
- Modulation of brain activity following botulinum toxin injection might suggest that botulinum toxin can remotely induce brain plasticity [2].

OBJECTIVE

To assess the modulation of oscillatory cortical activity after botulinum toxin injection in post-stroke patients.

METHODS

Participants

- 12 chronic post-stroke patients, able to actively extend the elbow with elbow flexors with muscle overactivity, without antispastic treatment for at least 4 months.

Injection procedure

- Patients received aboBoNT-A (abobotulinumtoxinA, Dysport, Ipsen Biopharm, Wrexham, UK) injected into one or more of the elbow flexor muscles.

Materials

- 64-channel ActiveTwo system (Biosemi) sampled at 1024 Hz.

Experimental task and design

- Non-controlled longitudinal prospective study
 - ULISCC-STROKE No ID-RCB: 2017-A01616-47
- Subjects performed two sets of ten active elbow extension at a self-spontaneous speed before aboBoNT-A injection (W0), 4 weeks (W4) and 16 weeks (W16) following the injection.

Data analysis

- Pre-processing was used to reduce the degree of artefact contamination consisting in: 1) channel rejection, 2) artefact subspace reconstruction, 3) independent component artefact rejection.
- Computing movement-related beta desynchronization (MRBD) corresponding to event-related power in the beta-band frequency (13-30 Hz).
- Effect of aboBoNT-A on MRBD over the time using a data-driven cluster-based permutation ANOVA at the topographical level.

RESULTS & DISCUSSION

- The analysis revealed **a main time effect over the sensorimotor cortices' electrodes (Fig 1.B)**. Post-hoc analyses revealed:

- A decrease in the cortical oscillatory activity localized in the sensorimotor cortices when comparing W4 to W0 (Fig. 1.C)
- An increase in the cortical oscillatory activity localized in the ipsilesional sensorimotor cortex when comparing W16 to W0 (Fig.1.D)

These results suggest:

- A **neuromodulatory effect** on the cortical oscillatory activity induced by aboBoNT-A injection, potentially explained by a **reduction of the sensorimotor afferent signal**.
- AboBoNT-A-induced plasticity after the injection characterized by increased activity in the ipsilesional cortex that could promote a **functional brain plasticity** [3].

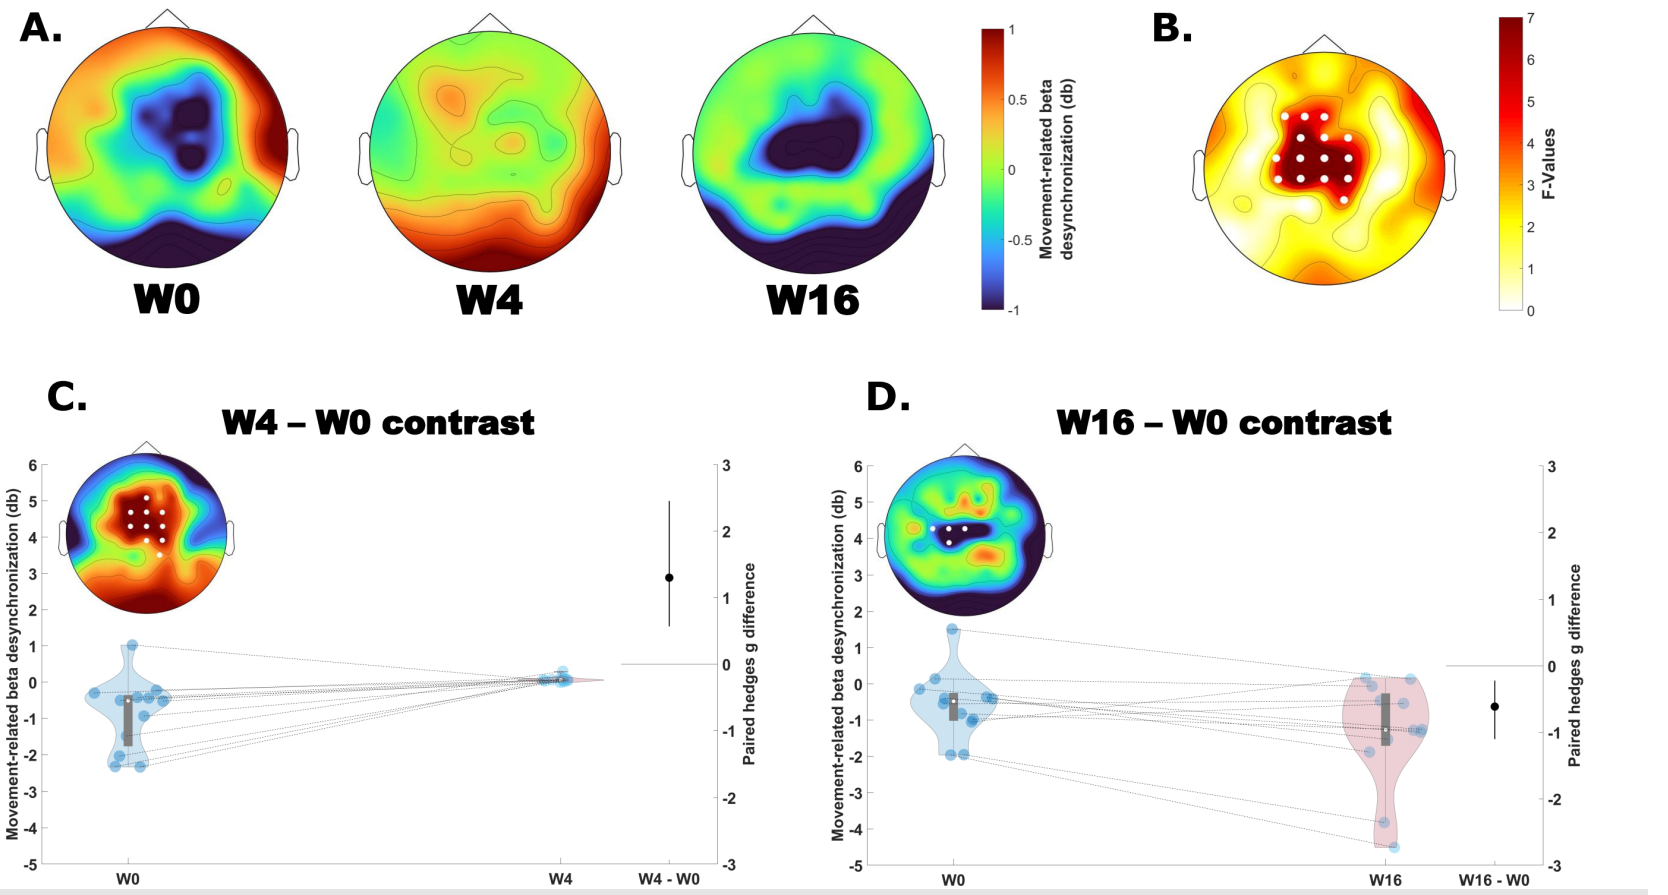
A.
W0
W4
W16
Movement-related beta desynchronization (db)
B.
F-Values
C.
W4 – W0 contrast
D.
W16 – W0 contrast
Movement-related beta desynchronization (db)
Paired hedges g difference
W0
W4
W4 - W0
W16
W16 - W0

Figure 1. A.) Topographic representation of MRBD at W0, W4 and W16. B.) Topographical plot representing the main time effect (white dots represent the significant time effect cluster) C.) Boxplot representing the significant difference between W4 and W0 on the MRBD over the identified significant cluster (white dots on the topographic plot represent the significant cluster over the W4-W0 contrast). D.) Boxplot representing the significant difference between W4 and W0 on the MRBD over the identified significant cluster (white dots on the topographic plot represent the significant cluster over the W16-W0 contrast)

CONCLUSIONS

Our results highlight :

- The potential role of aboBoNT-A injections combined with standard rehabilitation to promote brain plasticity.

- Longitudinal evaluation of cortical oscillatory activity could provide an understanding of the mechanisms of brain plasticity.

Disclosures: AC is employee of Ipsen Biopharma; JT, PM, DA, DG: None declared

Acknowledgements: The authors thank all patients involved in the study, as well as their caregivers, care team, investigators and research staff in participating institutions.

Presented at SOFAMEA 2020, 22-24 Janvier 2020, Nice, France

References :

[1] Wissel, J.et al. (2009). European consensus table on the use of botulinum toxin type A in adult spasticity. *Journal of Rehabilitation Medicine*, *41*(1), 13-25.
[2] Veverka, T. et al (2016). Sensorimotor modulation by botulinum toxin A in post-stroke arm spasticity: Passive hand movement. *Journal of the Neurological Sciences*, *362*, 14-20.
[3] Pelicioni, M. C. X et al (2016). *Functional versus Nonfunctional Rehabilitation in Chronic Ischemic Stroke: Evidences from a Randomized Functional MRI Study*. Neural Plasticity.